A Unified Platform for Rapid Pathogen Detection, Species Identification, and Antimicrobial Resistance Mutation Profiling

Jiaobao Song, Qingge Li *

** School of Life Sciences, Xiamen University - Xiamen (China)*

RES-243

INTRO.

The complex etiology in critically ill patients warrants prompt response harnessed with an accessible and affordable diagnostic platform that enables pathogen identification, species/type discrimination and drug resistance status determination. While sequencing platforms have found wide applications in research settings, their use in intensive care units is difficult in terms of both cost and time consumption.

MATERIALS & METHODS

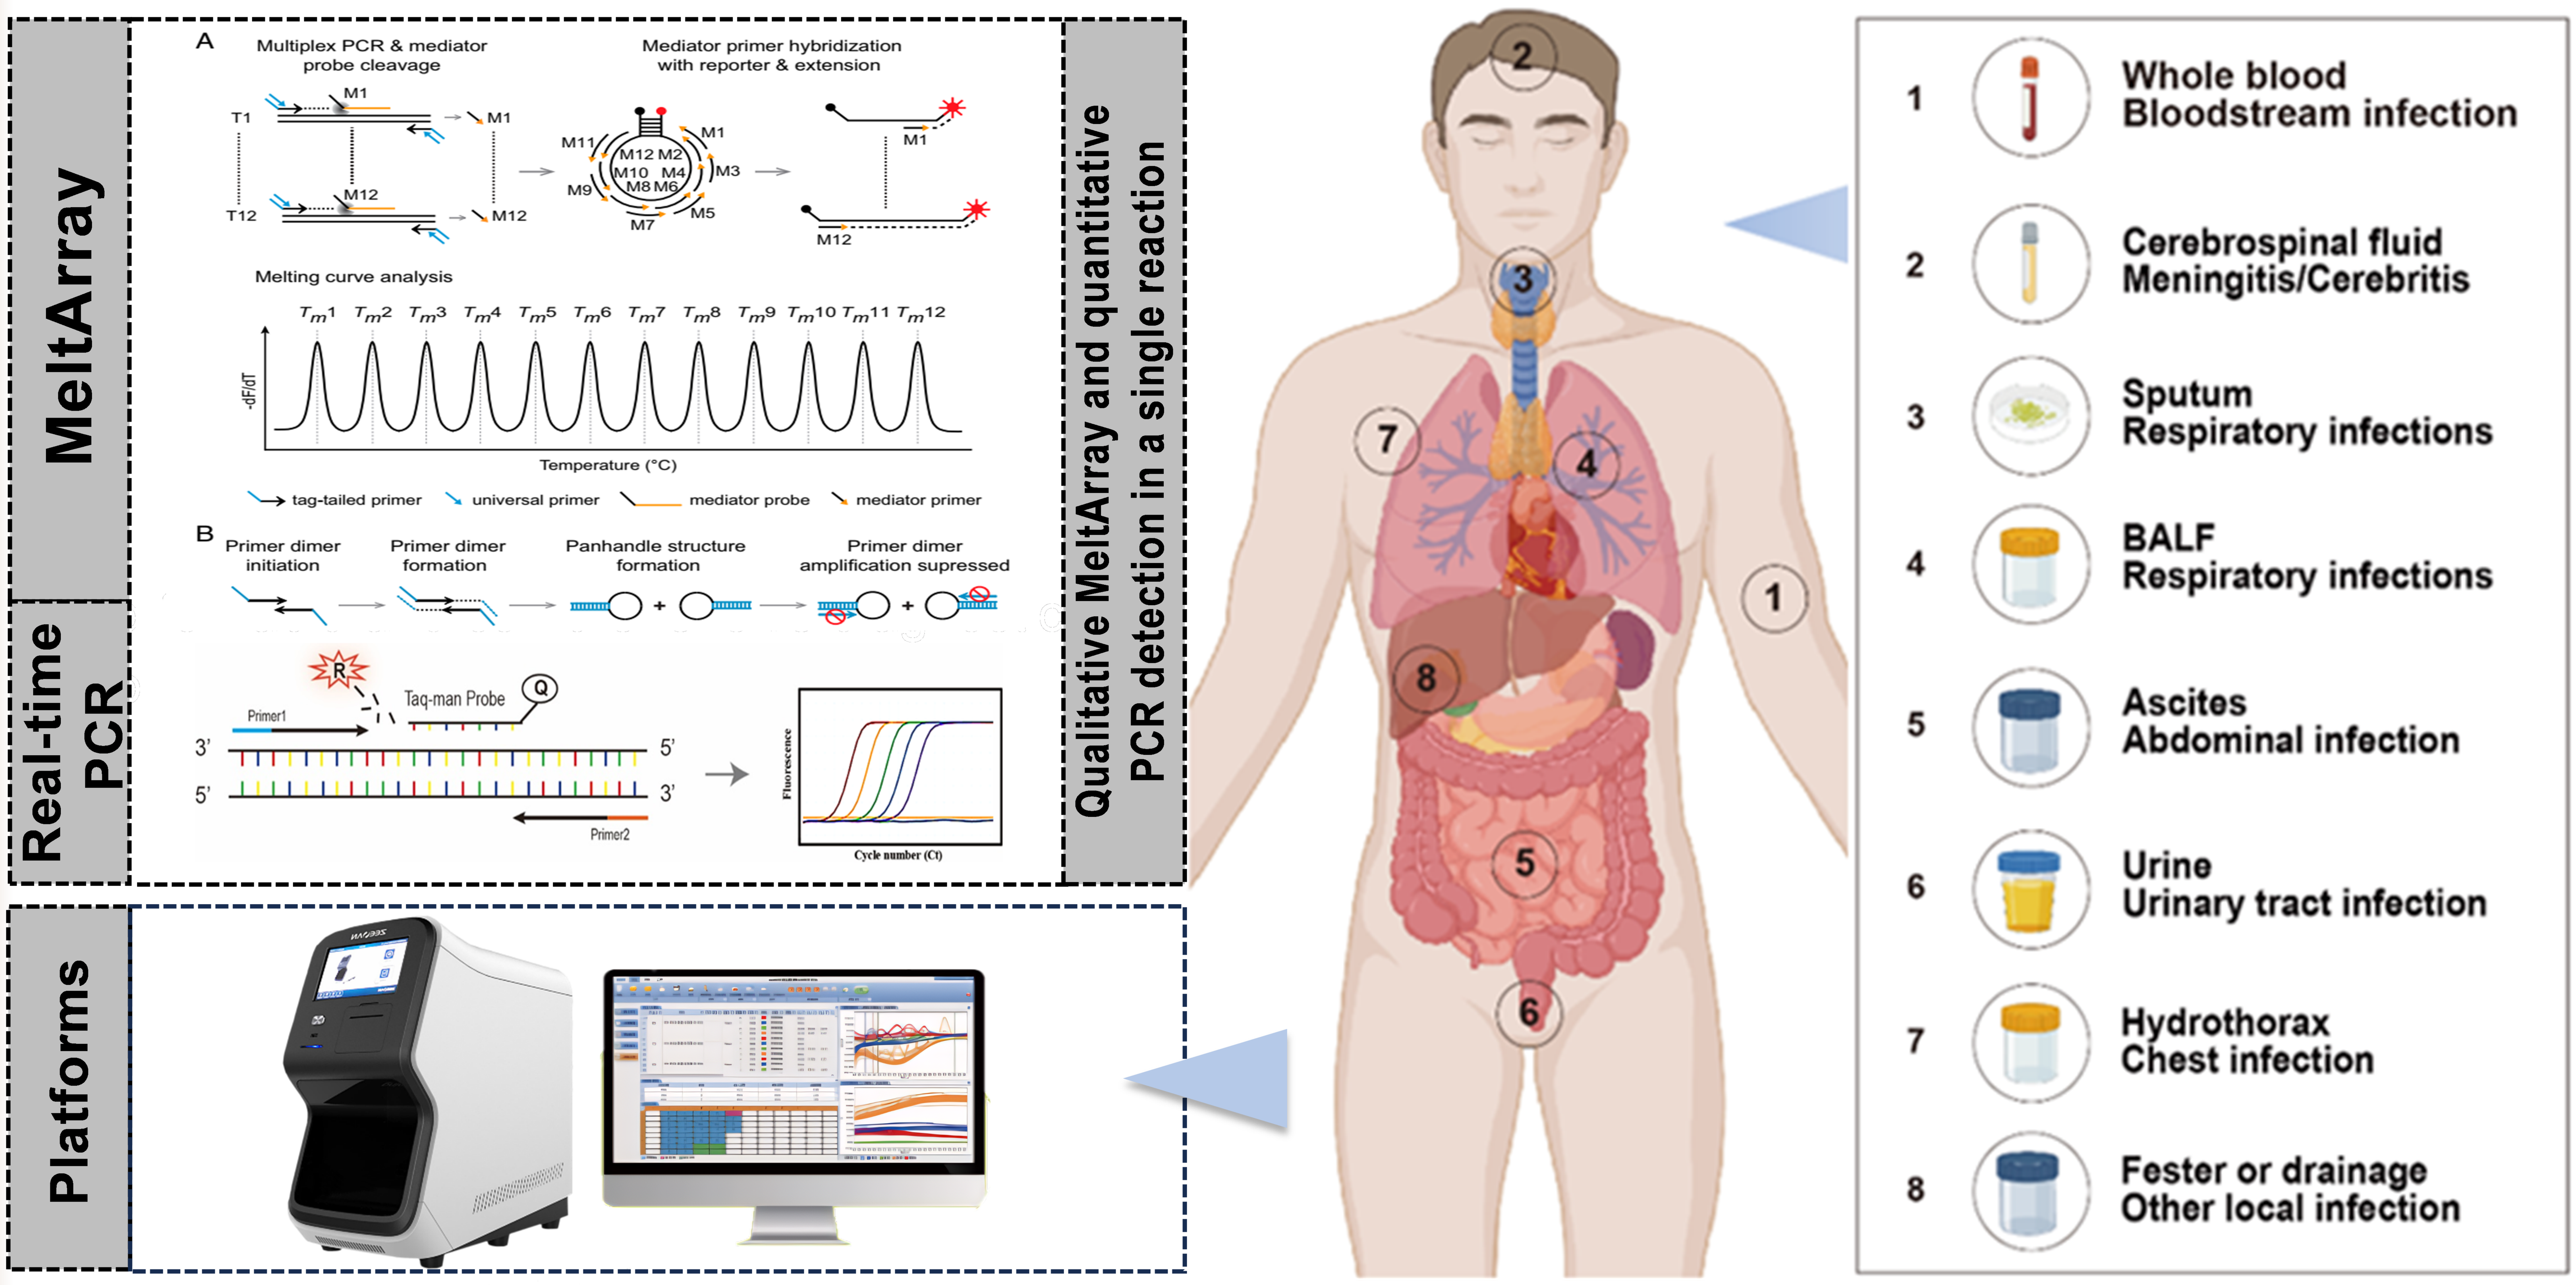
MeltArray
Real-time PCR
Platforms
Qualitative MeltArray and quantitative PCR detection in a single reaction
A
Multiplex PCR & mediator probe cleavage
Mediator primer hybridization with reporter & extension
Melting curve analysis
Temperature (°C)
tag-tailed primer
universal primer
mediator probe
mediator primer
B
Primer dimer initiation
Primer dimer formation
Panhandle structure formation
Primer dimer amplification supressed
Taq-man Probe
Primer1
Primer2
Fluorescence
Cycle number (Ct)
1 Whole blood Bloodstream infection
2 Cerebrospinal fluid Meningitis/Cerebritis
3 Sputum Respiratory infections
4 BALF Respiratory infections
5 Ascites Abdominal infection
6 Urine Urinary tract infection
7 Hydrothorax Chest infection
8 Fester or drainage Other local infection

RESULTS & DISCUSSION

ID of pathogen & AMR in ICU patients

- 3 Rxn: 62 pathogens
- 5 Rxn: 98 AMR genes

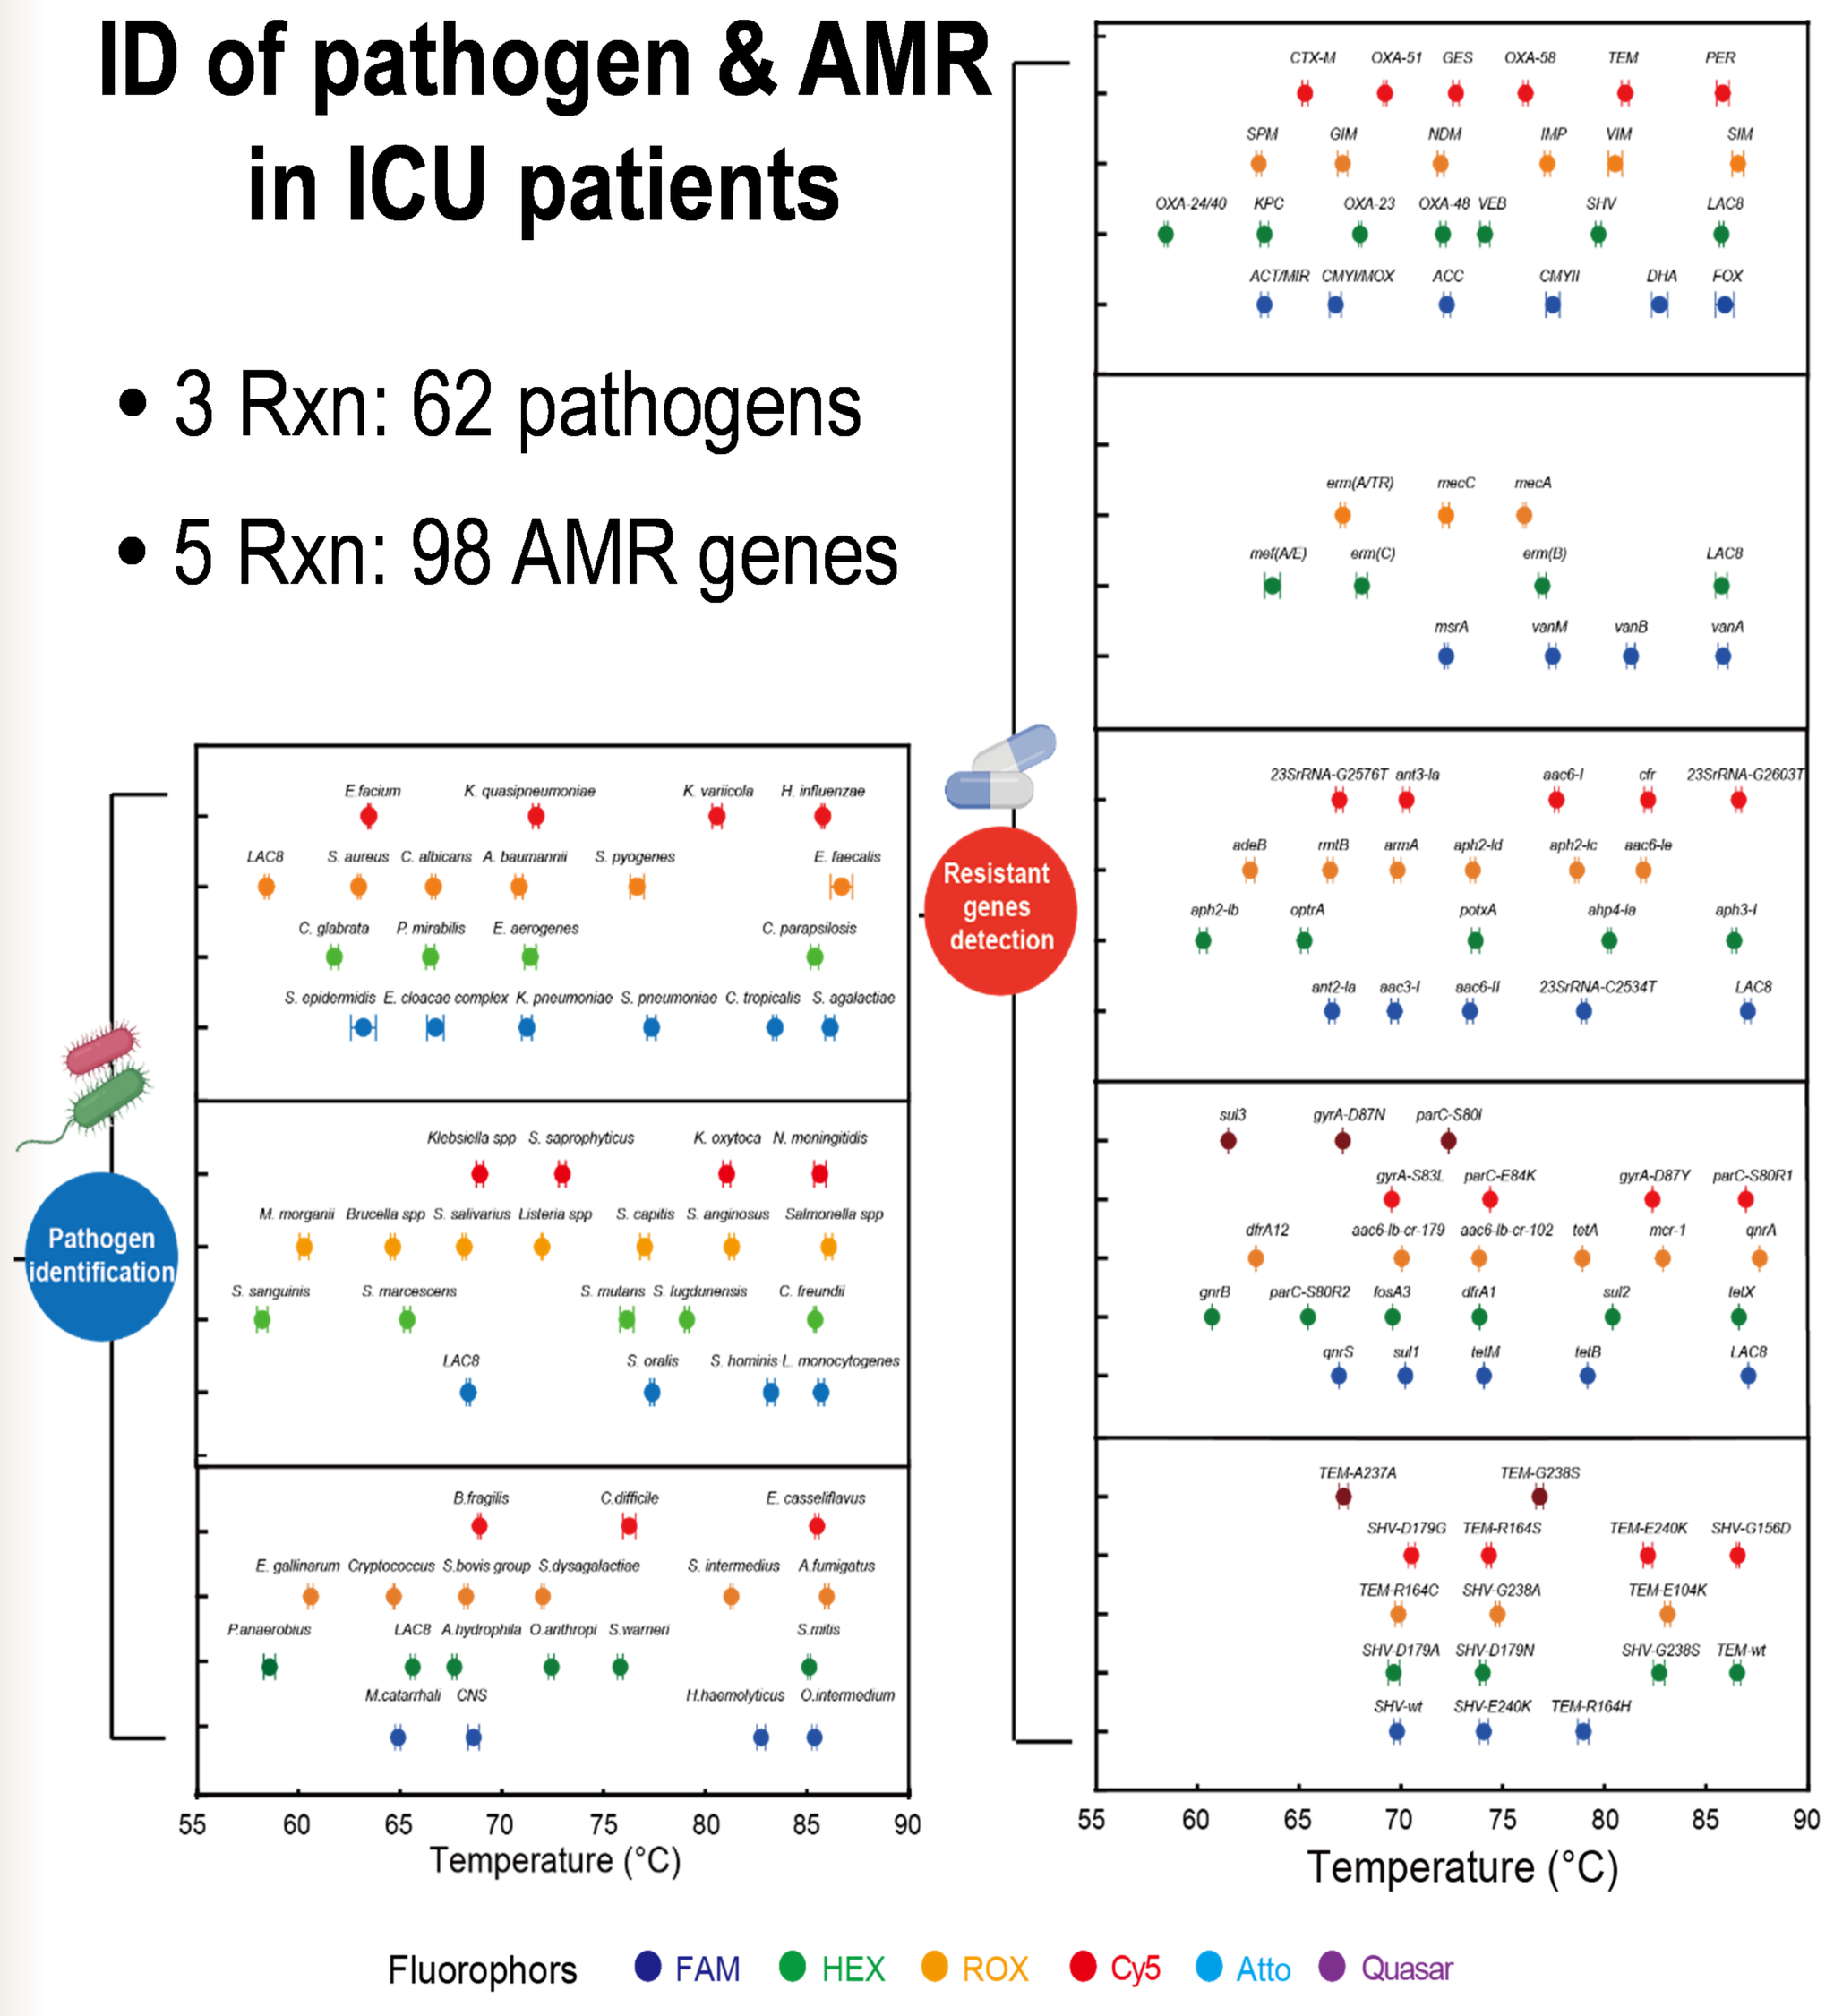
Pathogen identification
Resistant genes detection
Temperature (°C)
Fluorophors FAM HEX ROX Cy5 Atto Quasar

Fig 1. A united pathogens and AMR genes detection platform.

Detection flowchart and sample types

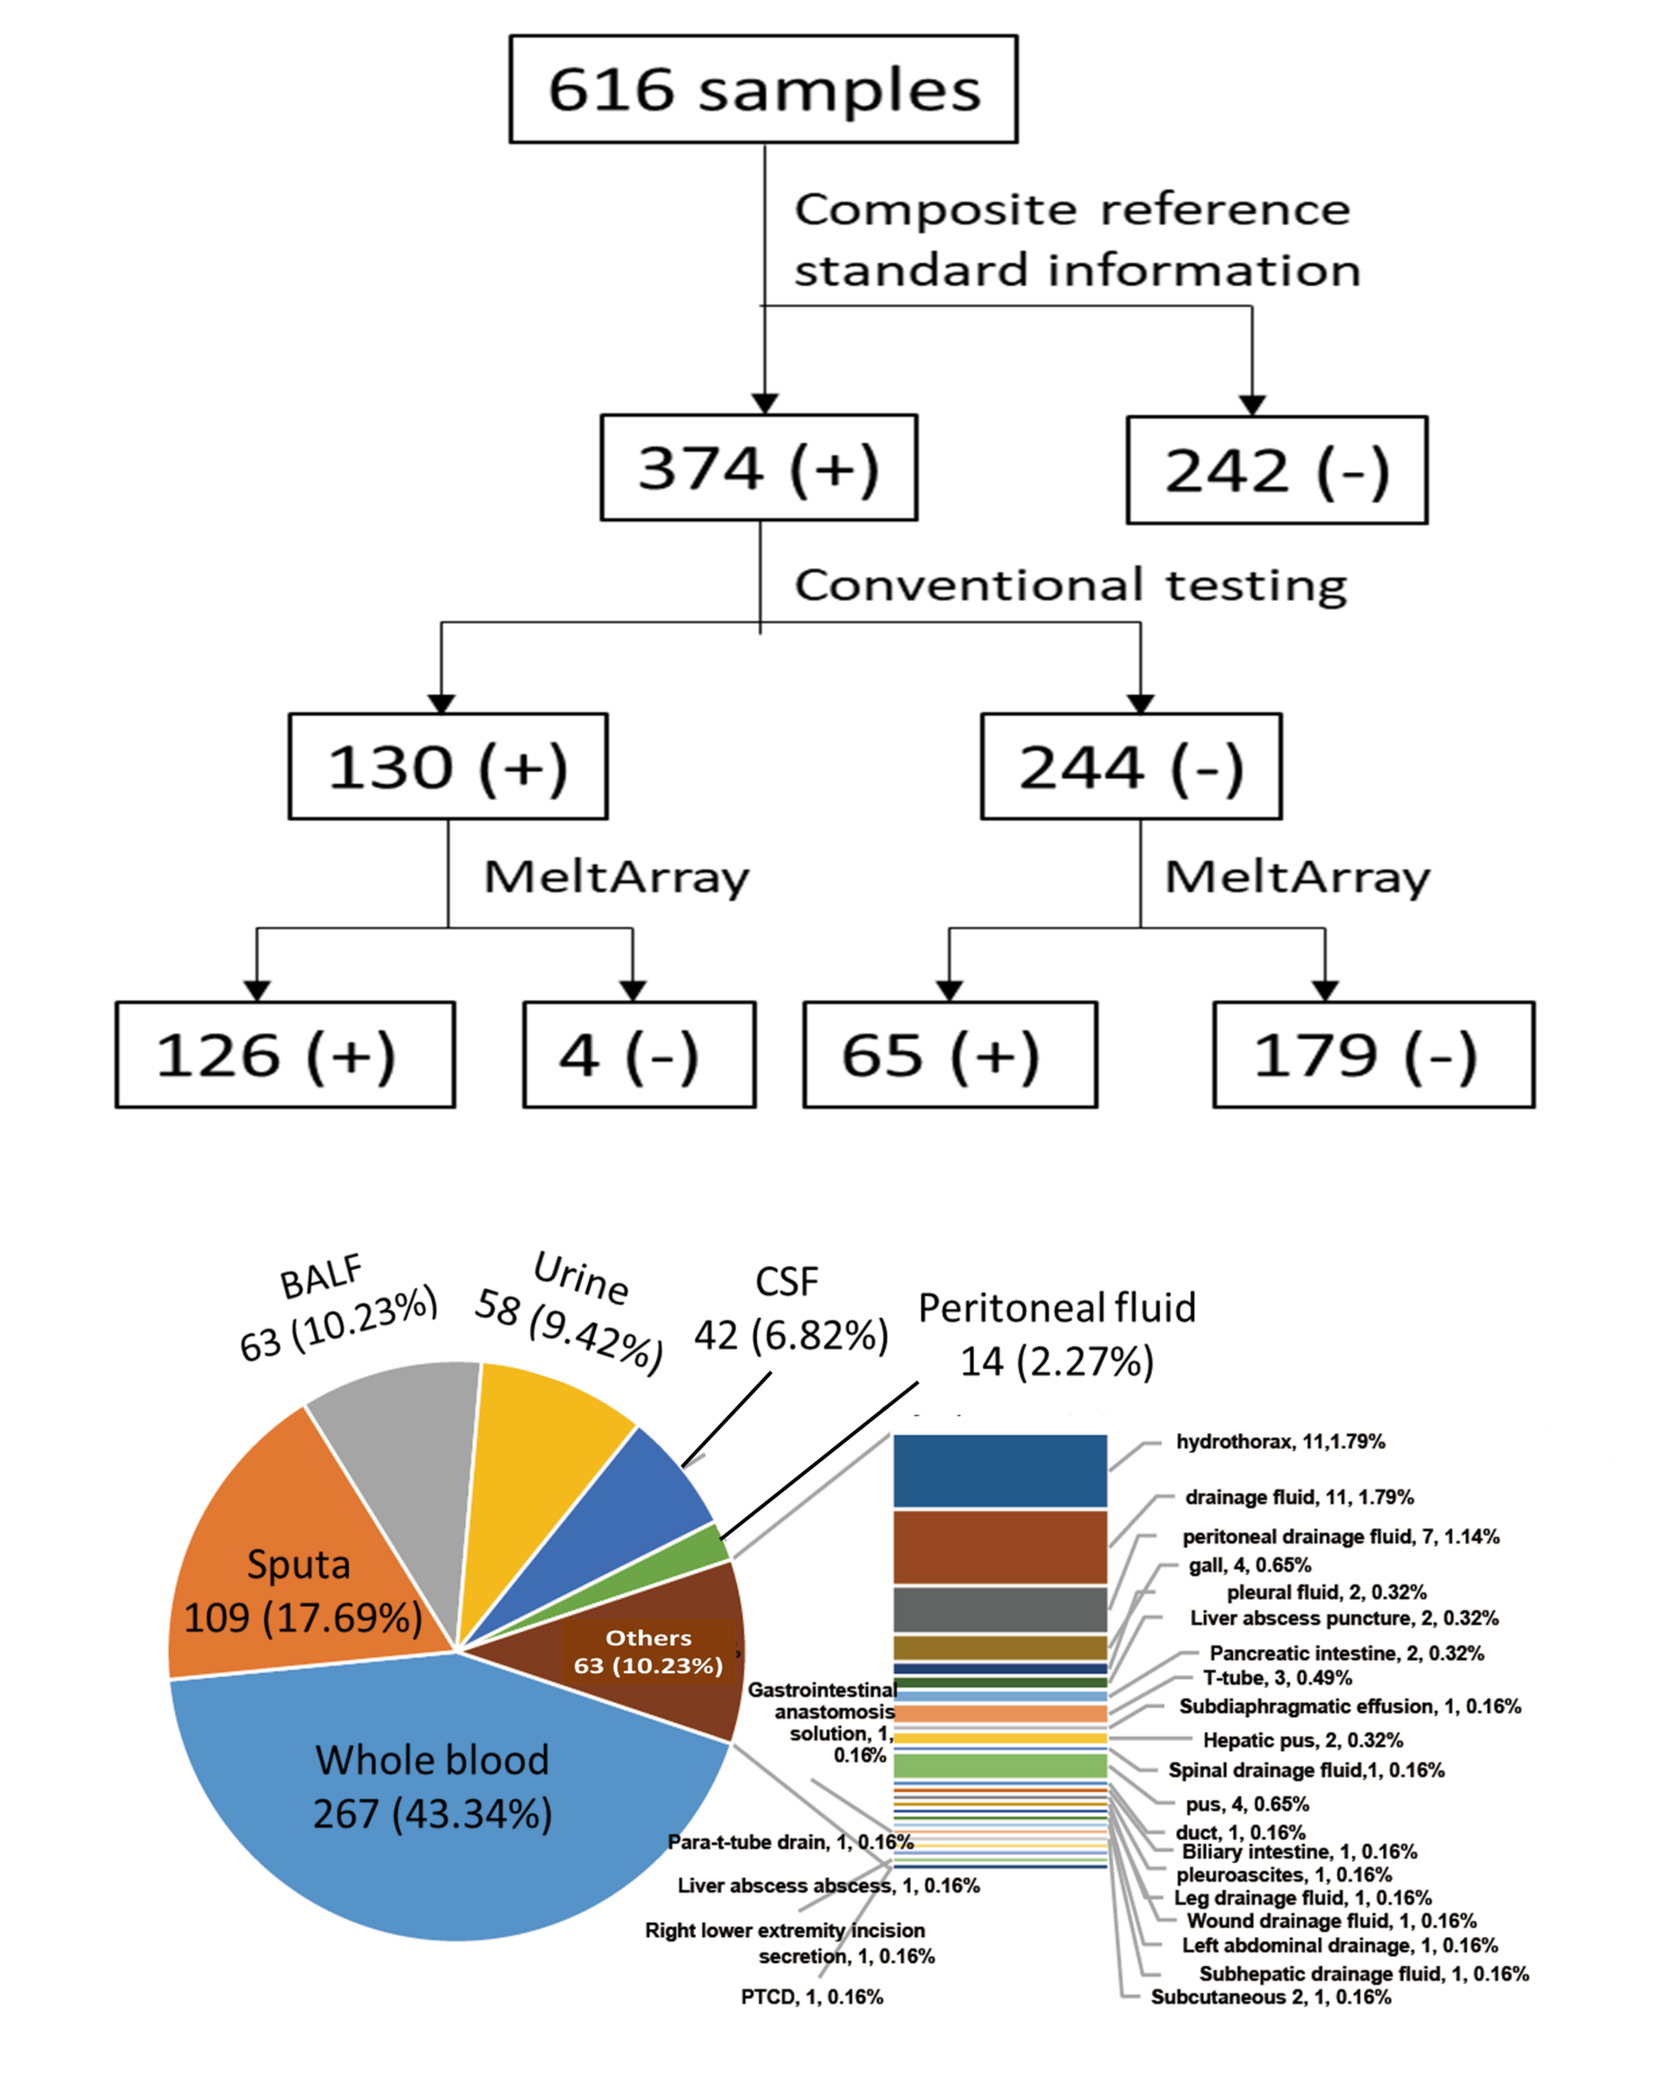
616 samples
Composite reference standard information
374 (+)
242 (-)
Conventional testing
130 (+)
244 (-)
MeltArray
126 (+)
4 (-)
65 (+)
179 (-)
BALF 63 (10.23%)
Urine 58 (9.42%)
CSF 42 (6.82%)
Peritoneal fluid 14 (2.27%)
Sputa 109 (17.69%)
Whole blood 267 (43.34%)
Others 63 (10.23%)

Fig 2. Clinical evaluation of MeltArray with ICU samples.

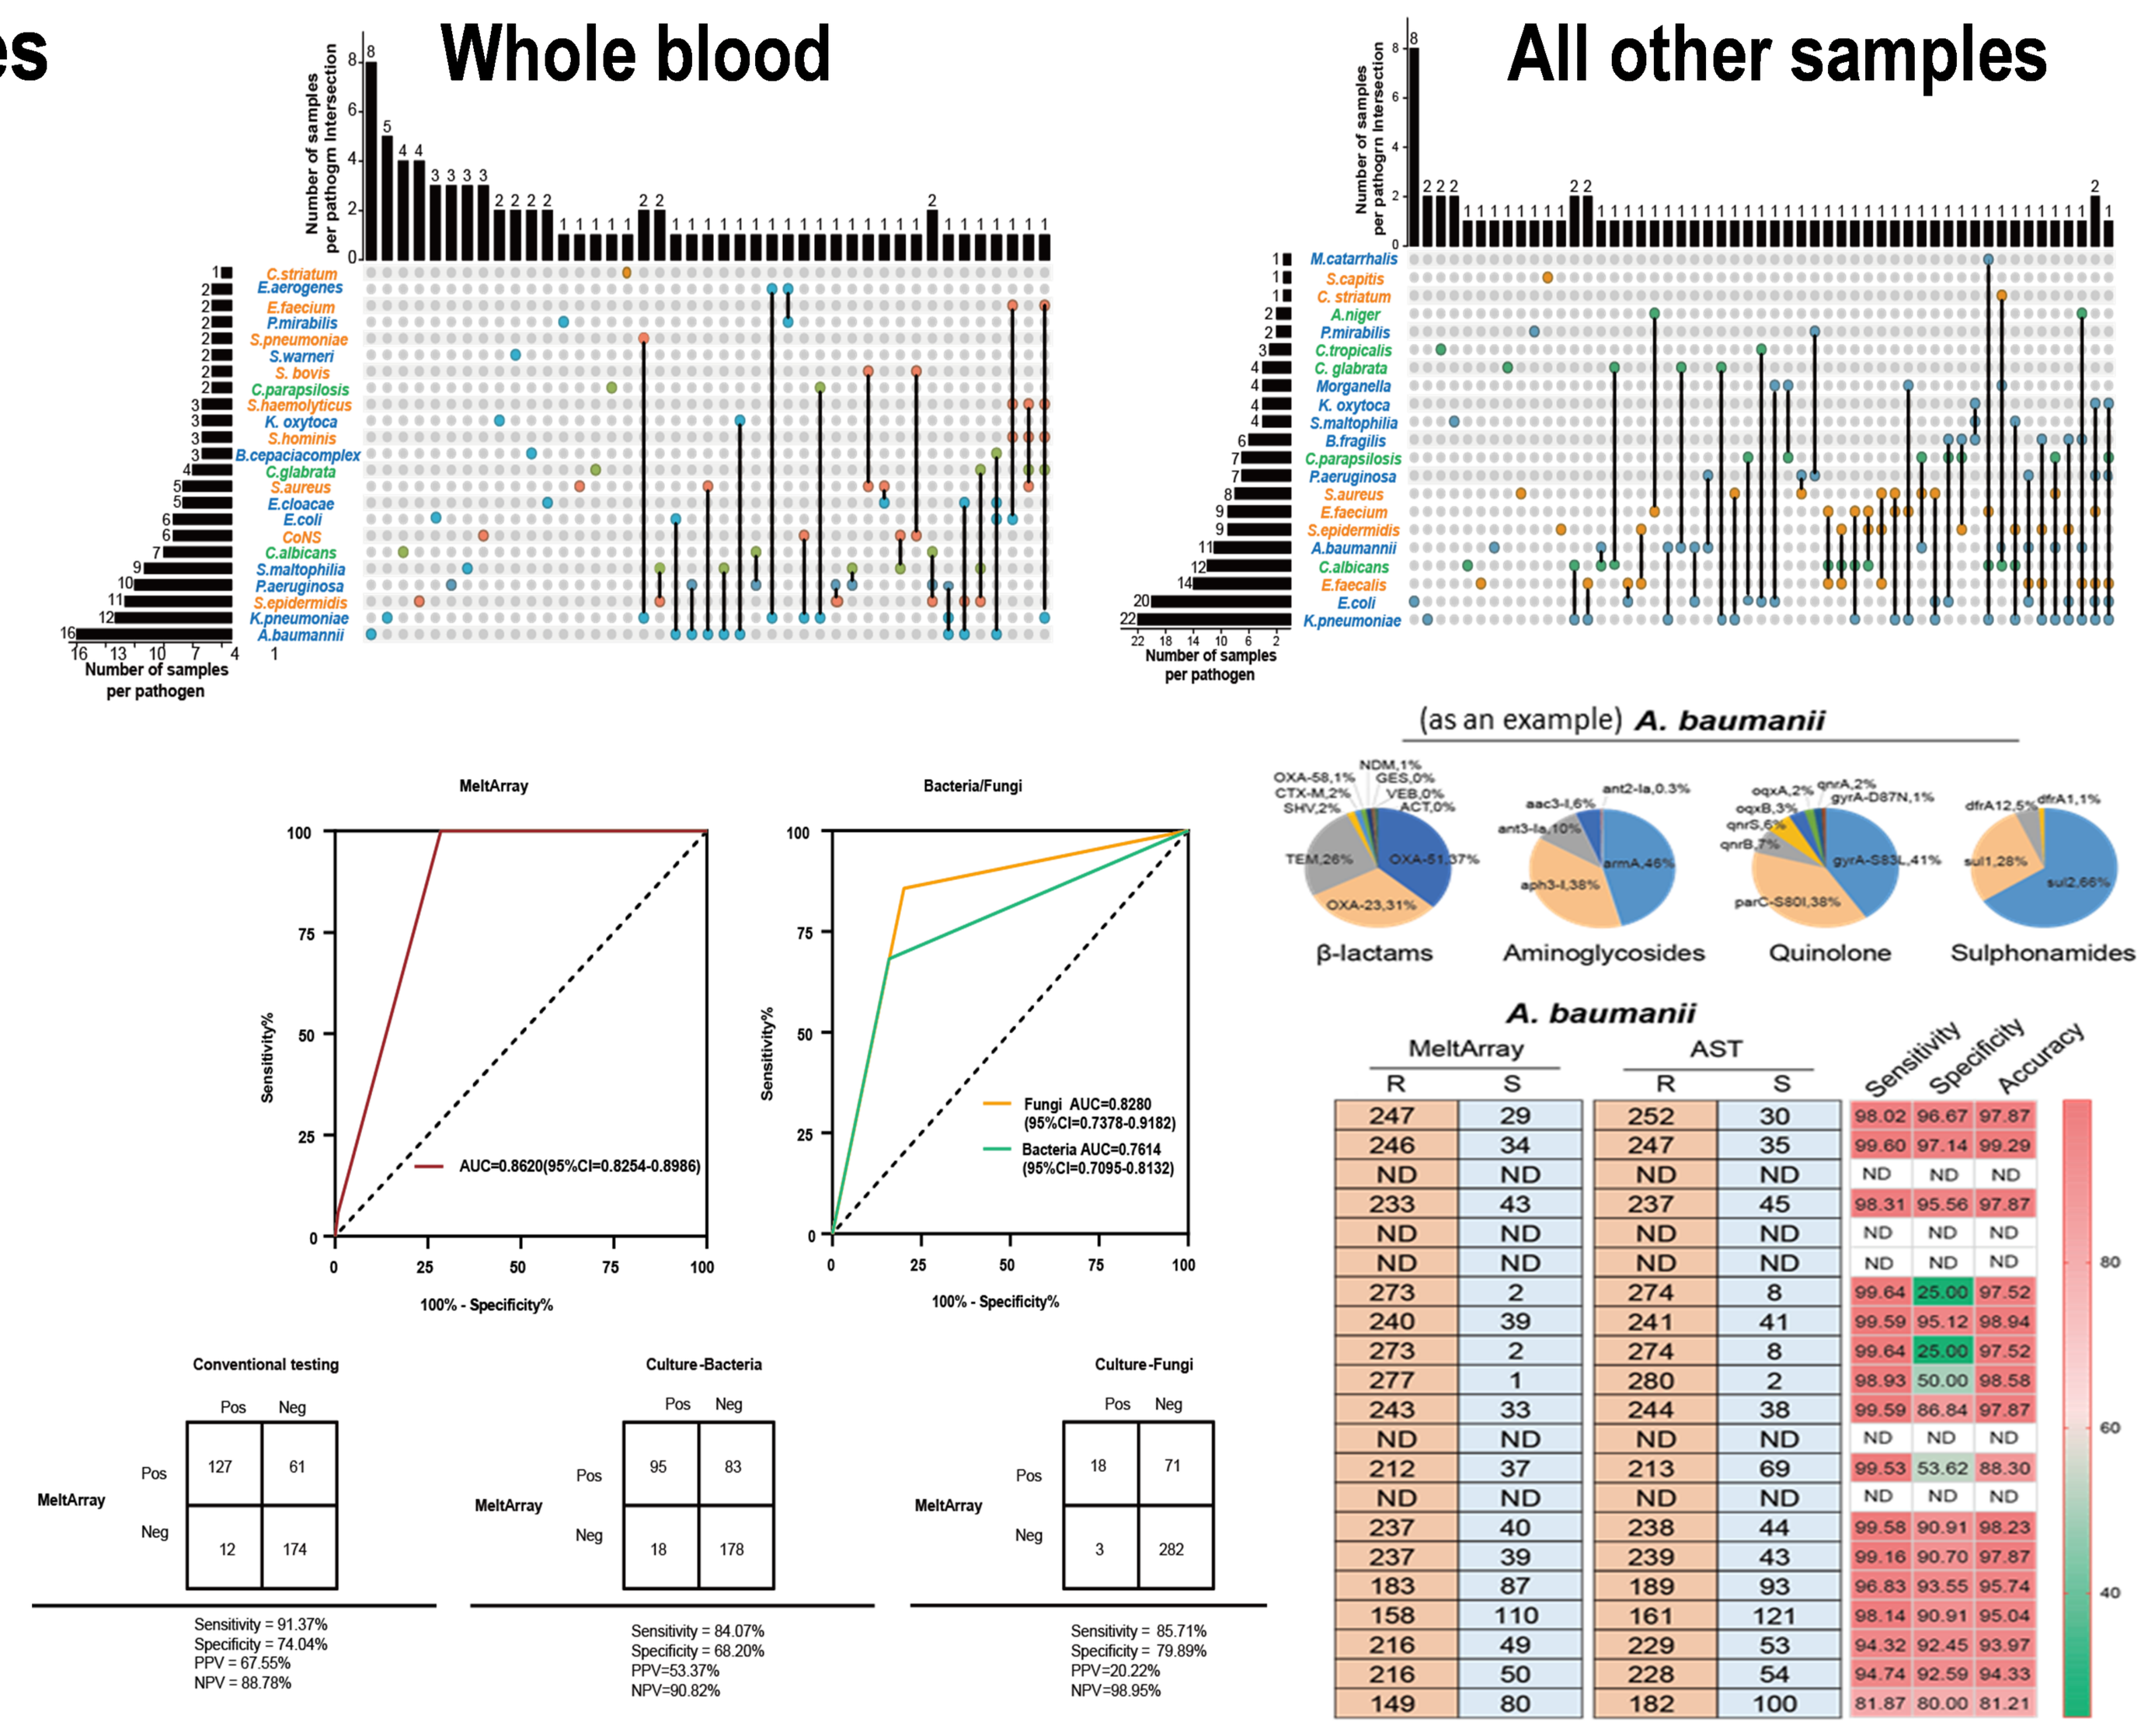
Whole blood
All other samples
Number of samples per pathogen
MeltArray
Bacteria/Fungi
AUC=0.8620(95%CI=0.8254-0.8986)
Fungi AUC=0.8280 (95%CI=0.7378-0.9182)
Bacteria AUC=0.7614 (95%CI=0.7095-0.8132)
Conventional testing
Sensitivity = 91.37%
Specificity = 74.04%
PPV = 67.55%
NPV = 88.78%
Culture-Bacteria
Sensitivity = 84.07%
Specificity = 68.20%
PPV=53.37%
NPV=90.82%
Culture-Fungi
Sensitivity = 85.71%
Specificity = 79.89%
PPV=20.22%
NPV=98.95%
(as an example) A. baumanii
β-lactams
Aminoglycosides
Quinolone
Sulphonamides

A. baumanii

MeltArray R	MeltArray S	AST R	AST S	Sensitivity	Specificity	Accuracy
247	29	252	30	98.02	96.67	97.87
246	34	247	35	99.60	97.14	99.29
ND	ND	ND	ND	ND	ND	ND
233	43	237	45	98.31	95.56	97.87
ND	ND	ND	ND	ND	ND	ND
ND	ND	ND	ND	ND	ND	ND
273	2	274	8	99.64	25.00	97.52
240	39	241	41	99.59	95.12	98.94
273	2	274	8	99.64	25.00	97.52
277	1	280	2	98.93	50.00	98.58
243	33	244	38	99.59	86.84	97.87
ND	ND	ND	ND	ND	ND	ND
212	37	213	69	99.53	53.62	88.30
ND	ND	ND	ND	ND	ND	ND
237	40	238	44	99.58	90.91	98.23
237	39	239	43	99.16	90.70	97.87
183	87	189	93	96.83	93.55	95.74
158	110	161	121	98.14	90.91	95.04
216	49	229	53	94.32	92.45	93.97
216	50	228	54	94.74	92.59	94.33
149	80	182	100	81.87	80.00	81.21

Fig 3. MeltArray vs conventional testing and drug susceptibility testing.

CONC.

Compared with current platform, MeltArray has the combined advantages of a short turnaround time, high sensitivity, wide pathogen coverage, and rapid antimicrobial resistance analysis. Implementation of the MeltArray approach may significantly improve current infectious diseases diagnosis and facilitate antibiotics stewardship programs.

Scan Me

APCCMI2025 BANGKOK

ACKNOWLEDGEMENTS: This study was supported by the NSFC Grant No. 82472368.

CONTACT: Li Qingge, PhD, Tel. 86-0592-2187363; Fax. 86-0592-2187363; Email: qgli@xmu.edu.cn

ZEESAN Biotech 致善生物