Closing Diagnostic Gaps in Respiratory Infections: A 40-Target Multiplex PCR Assay for Comprehensive Pathogen Detection

Wen-Tyng Kang [1], Qingge Li [2]

[1]Zeesan Biotech - Xiamen (China), [2]Xiamen University - Xiamen (China)

RES-235

INTRODUCTION

Respiratory tract infections (RTIs) remain a major global health burden, with **upper RTIs** accounting for more than **12.8 billion** cases annually and **lower RTIs** responsible for **2.4 million** deaths worldwide [1,2]. This burden is further compounded by the emergence of pathogens such as SARS-CoV-2, influenza A/B, and their novel variants, as well as the frequent occurrence of bacterial co-infections.

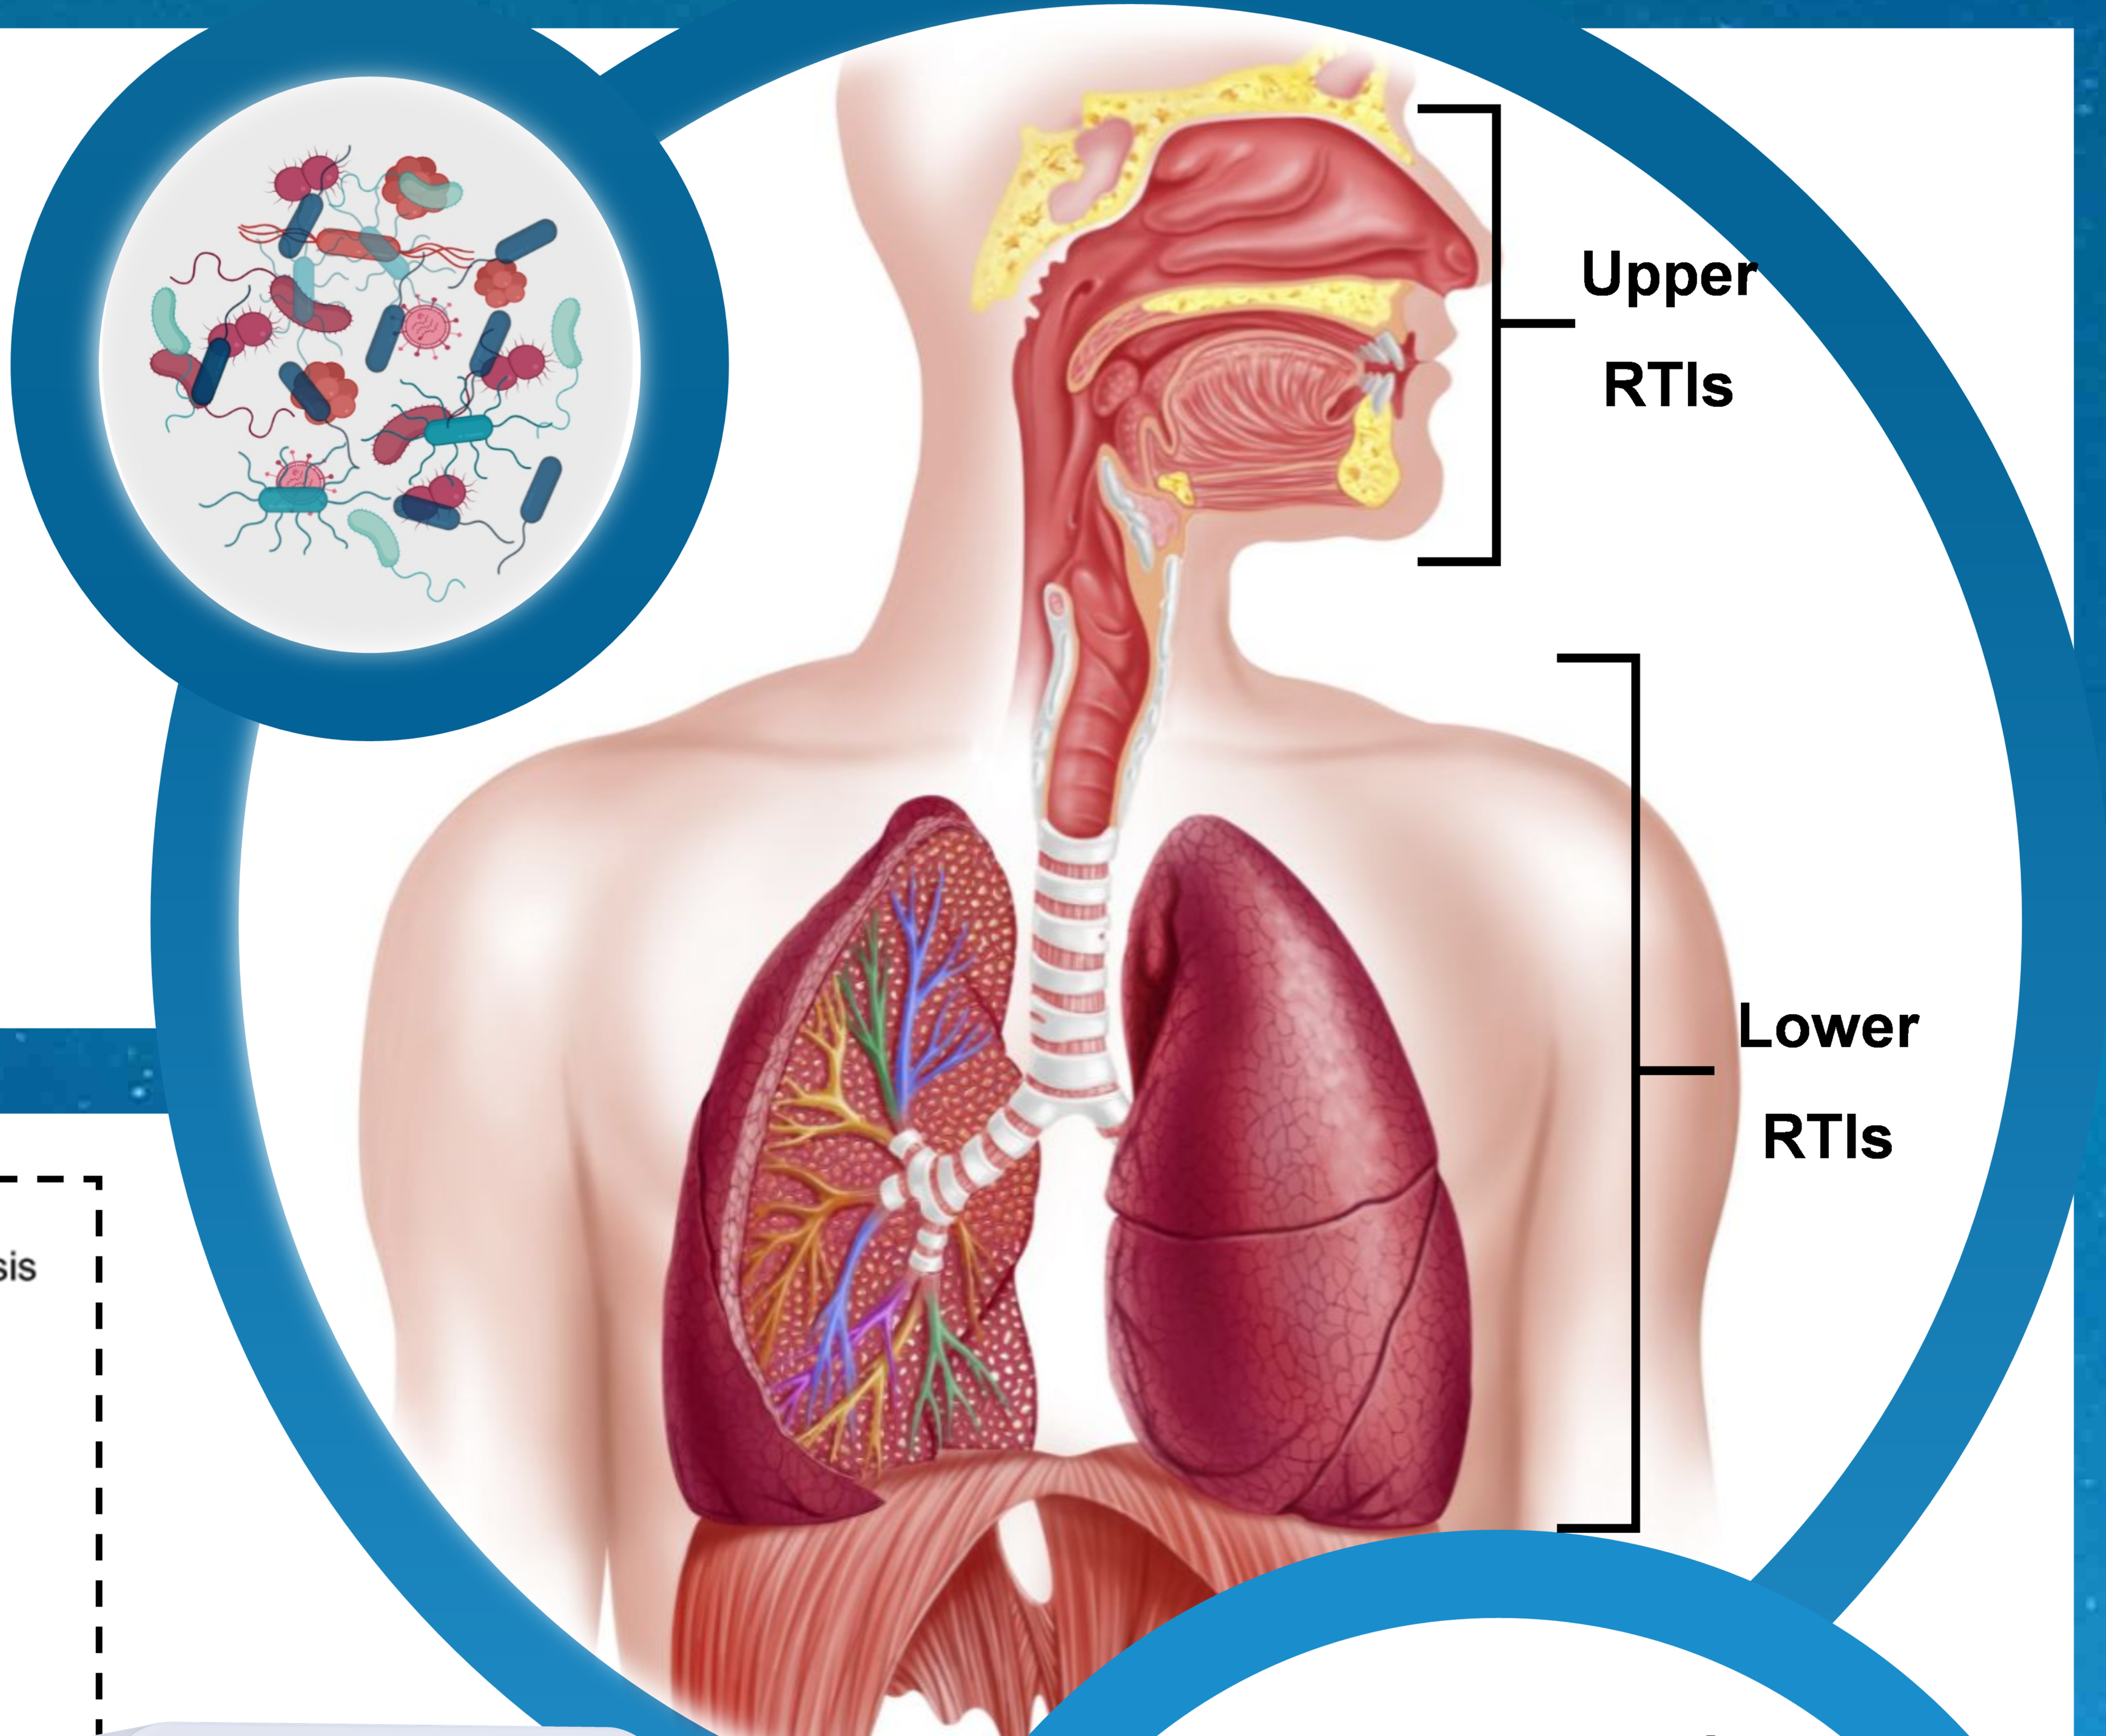
Upper
RTIs
Lower
RTIs

MATERIALS & METHODS

MeltArray

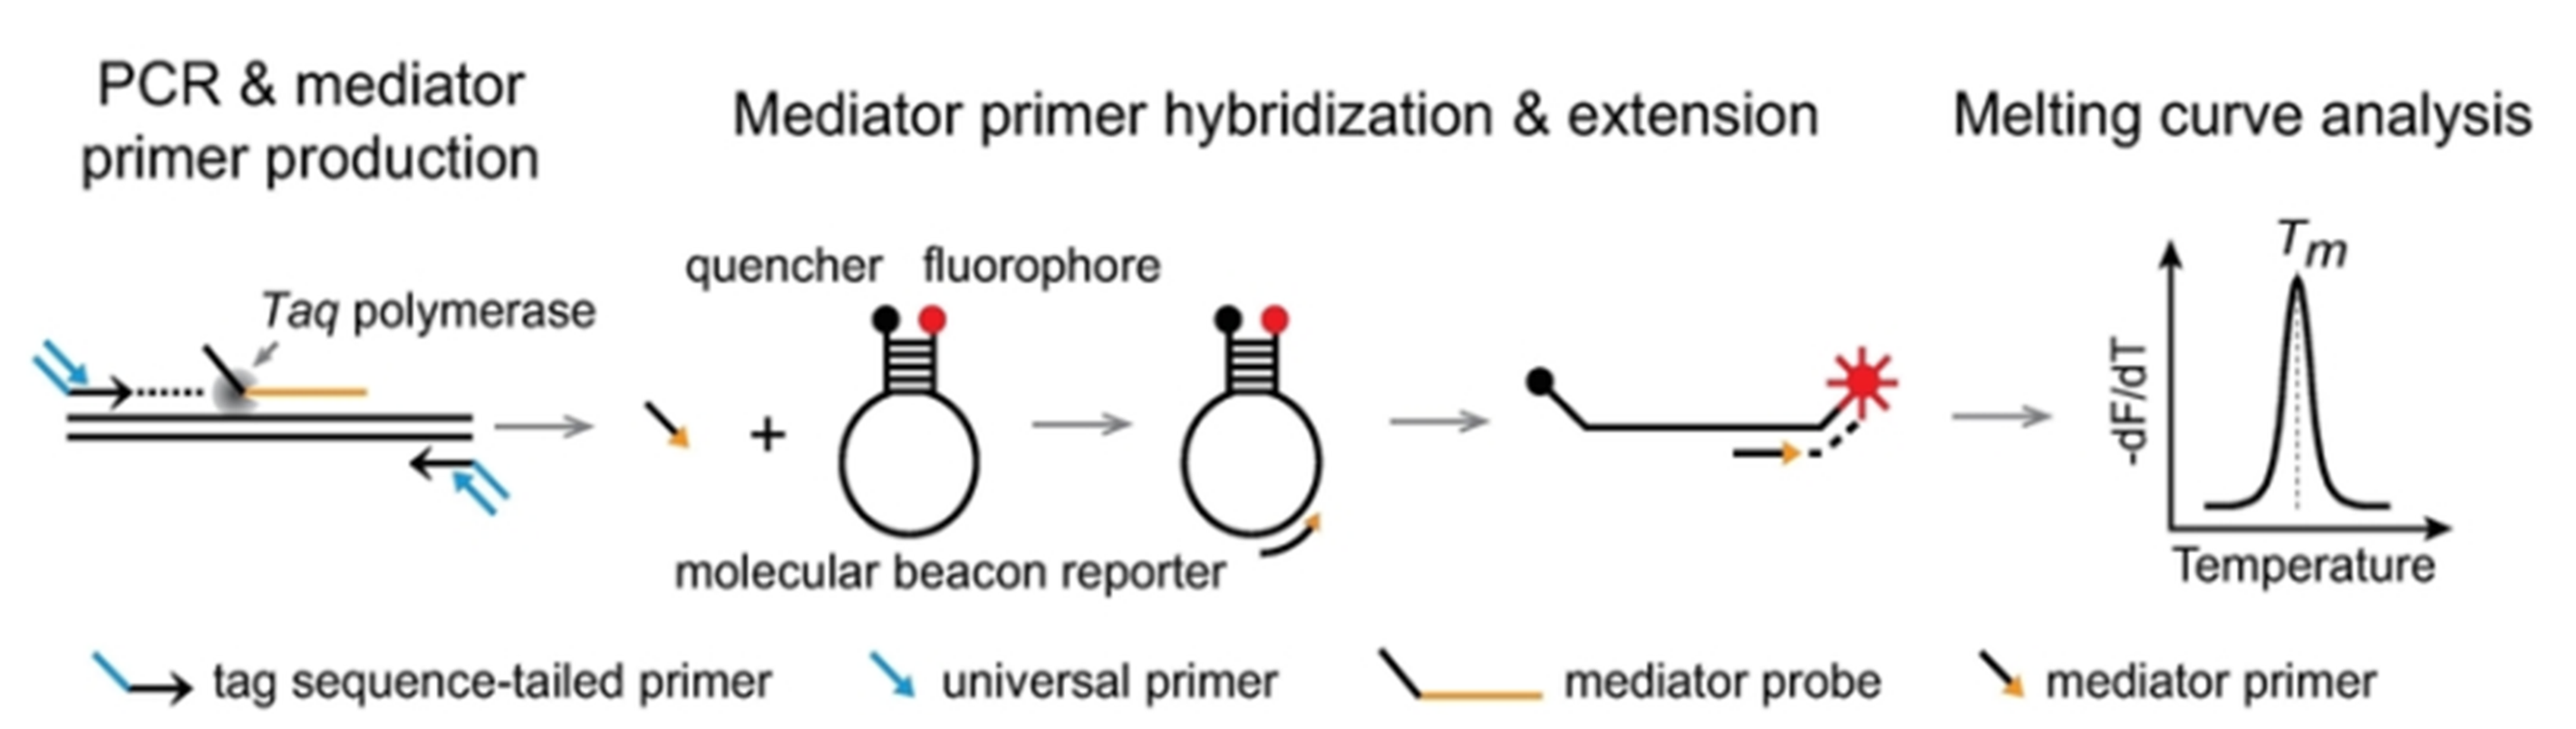
PCR & mediator primer production
Mediator primer hybridization & extension
Melting curve analysis
Taq polymerase
quencher
fluorophore
molecular beacon reporter
T_m
-dF/dT
Temperature
tag sequence-tailed primer
universal primer
mediator probe
mediator primer

Two-dimensional tag-based detection using a combination of fluorescence channels and melting temperatures [3].

Real-time PCR

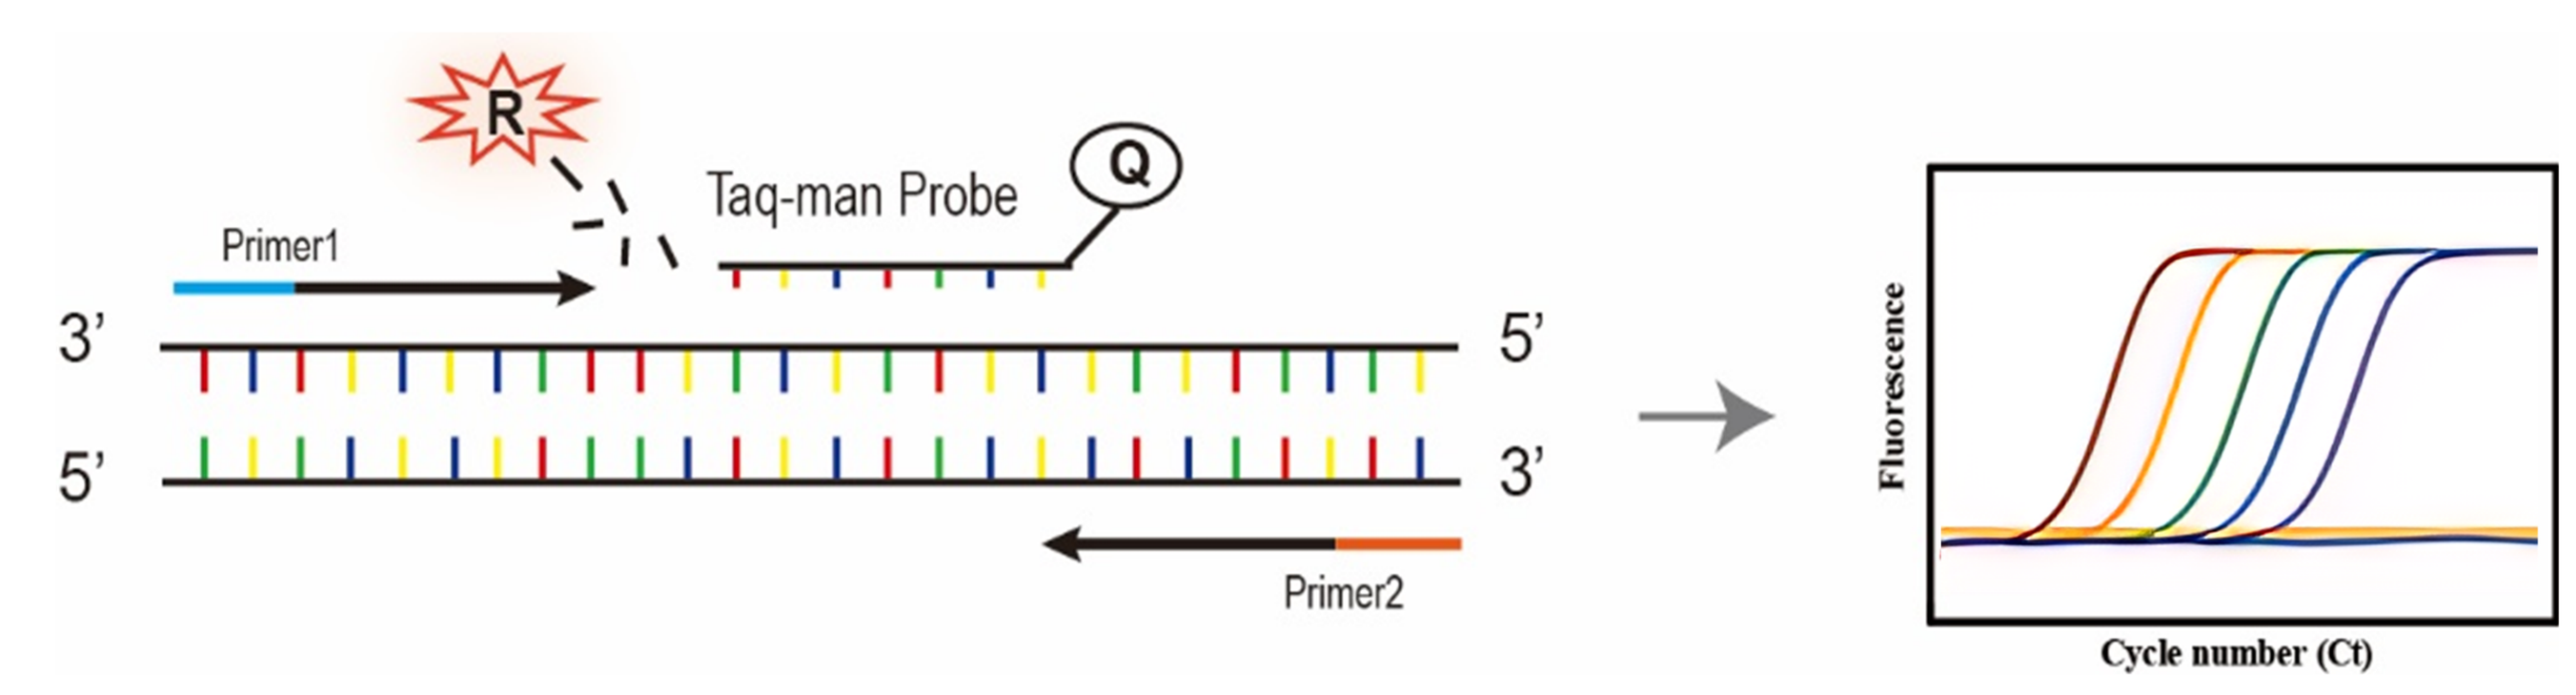
R
Taq-man Probe
Q
Primer1
3'
5'
5'
3'
Primer2
Fluorescence
Cycle number (Ct)

Quantitative detection based on qPCR technology using TaqMan probes.

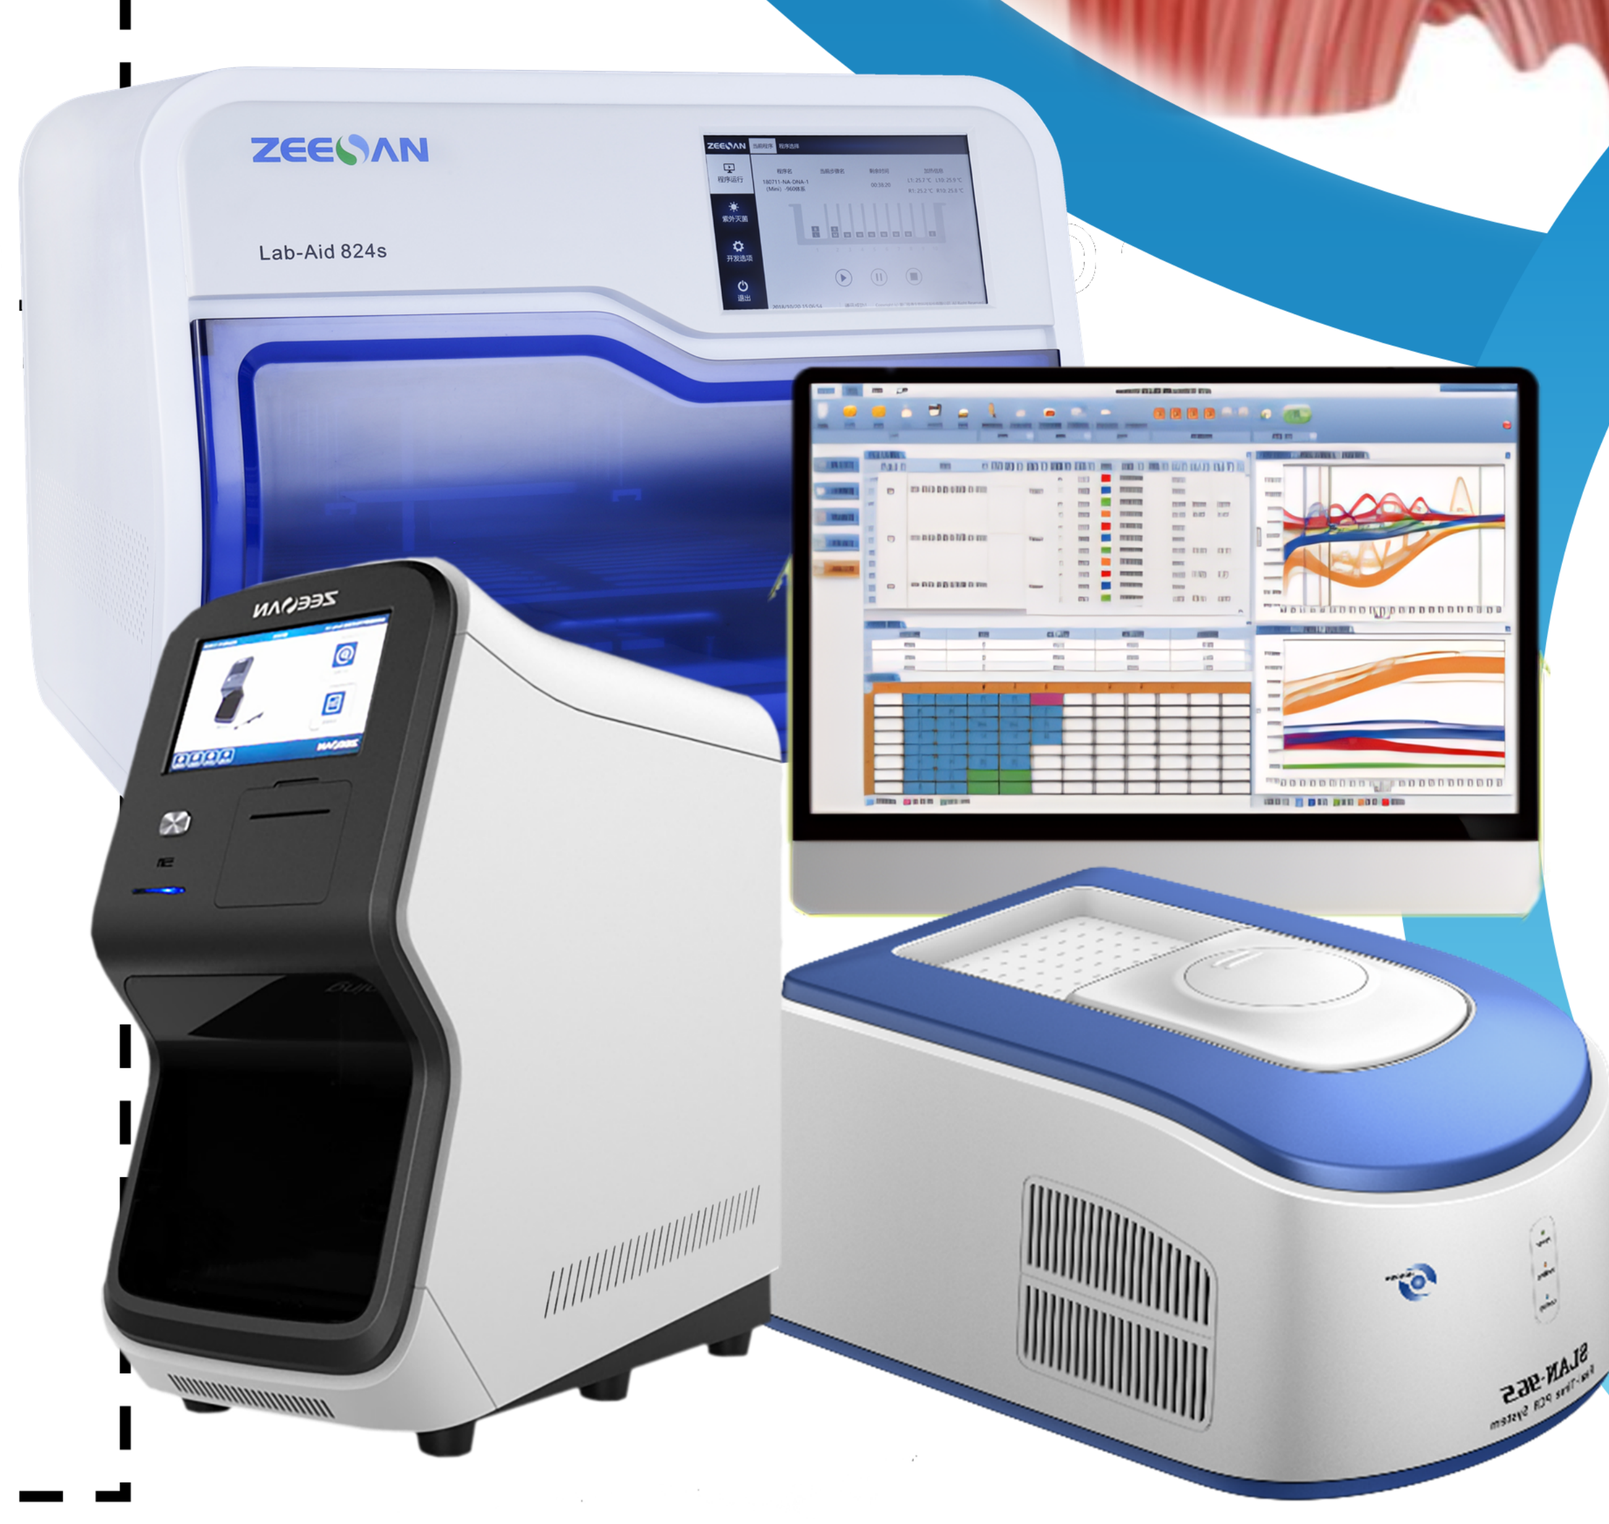
ZEESAN
Lab-Aid 824s

Urgent need for **rapid and comprehensive RTI diagnostics** to support clinical decision-making and antimicrobial stewardship.

RESULTS & DISCUSSION

- **Detects 32 pathogens** via unique fluorescent channels and T_m values.
- **Quantifies 8 bacterial** species by real-time PCR Ct values.
- Quantification is essential for assessing colonizing bacterial species in immuno-compromised patients.

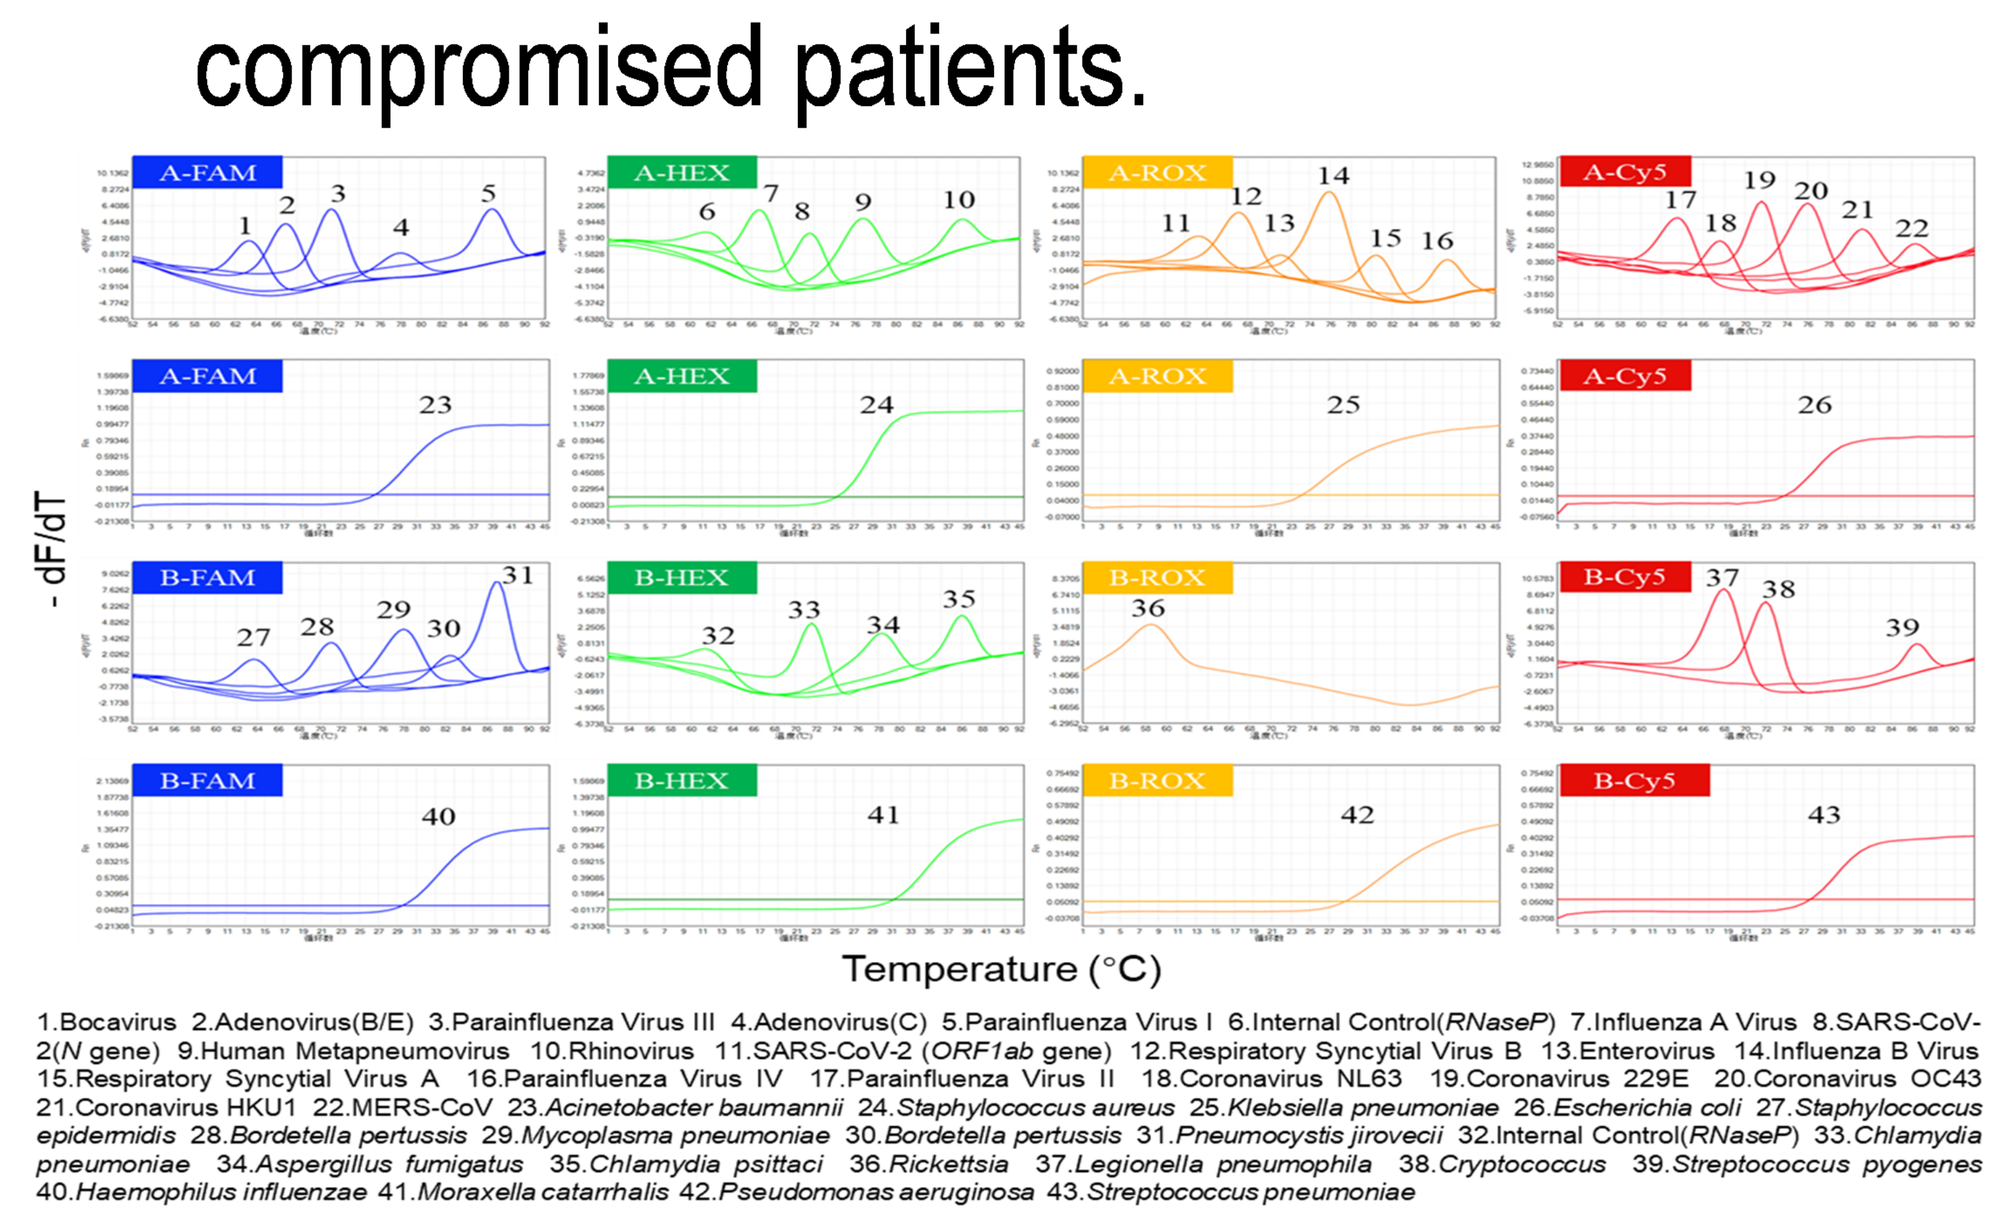
A-FAM
A-HEX
A-ROX
A-Cy5
B-FAM
B-HEX
B-ROX
B-Cy5
- dF/dT
Temperature (°C)
1.Bocavirus 2.Adenovirus(B/E) 3.Parainfluenza Virus III 4.Adenovirus(C) 5.Parainfluenza Virus I 6.Internal Control(RNaseP) 7.Influenza A Virus 8.SARS-CoV-2(N gene) 9.Human Metapneumovirus 10.Rhinovirus 11.SARS-CoV-2 (ORF1ab gene) 12.Respiratory Syncytial Virus B 13.Enterovirus 14.Influenza B Virus 15.Respiratory Syncytial Virus A 16.Parainfluenza Virus IV 17.Parainfluenza Virus II 18.Coronavirus NL63 19.Coronavirus 229E 20.Coronavirus OC43 21.Coronavirus HKU1 22.MERS-CoV 23.Acinetobacter baumannii 24.Staphylococcus aureus 25.Klebsiella pneumoniae 26.Escherichia coli 27.Staphylococcus epidermidis 28.Bordetella pertussis 29.Mycoplasma pneumoniae 30.Bordetella pertussis 31.Pneumocystis jirovecii 32.Internal Control(RNaseP) 33.Chlamydia pneumoniae 34.Aspergillus fumigatus 35.Chlamydia psittaci 36.Rickettsia 37.Legionella pneumophila 38.Cryptococcus 39.Streptococcus pyogenes 40.Haemophilus influenzae 41.Moraxella catarrhalis 42.Pseudomonas aeruginosa 43.Streptococcus pneumoniae

Figure 1. Typical result of RPP-40 for MeltArray and real-time PCR.

- **Clinical trial:** 1,036 oropharyngeal (OP) swabs and 3 simulated swabs (MERS-CoV diluted in negatives).
- RPP-40 assay **fully automated on SLAN qPCR / Sanity 2.0s**.
- Results in **≤2.5 h**, minimal hands-on time.
- LOD **<500 copies/mL** for all targets.

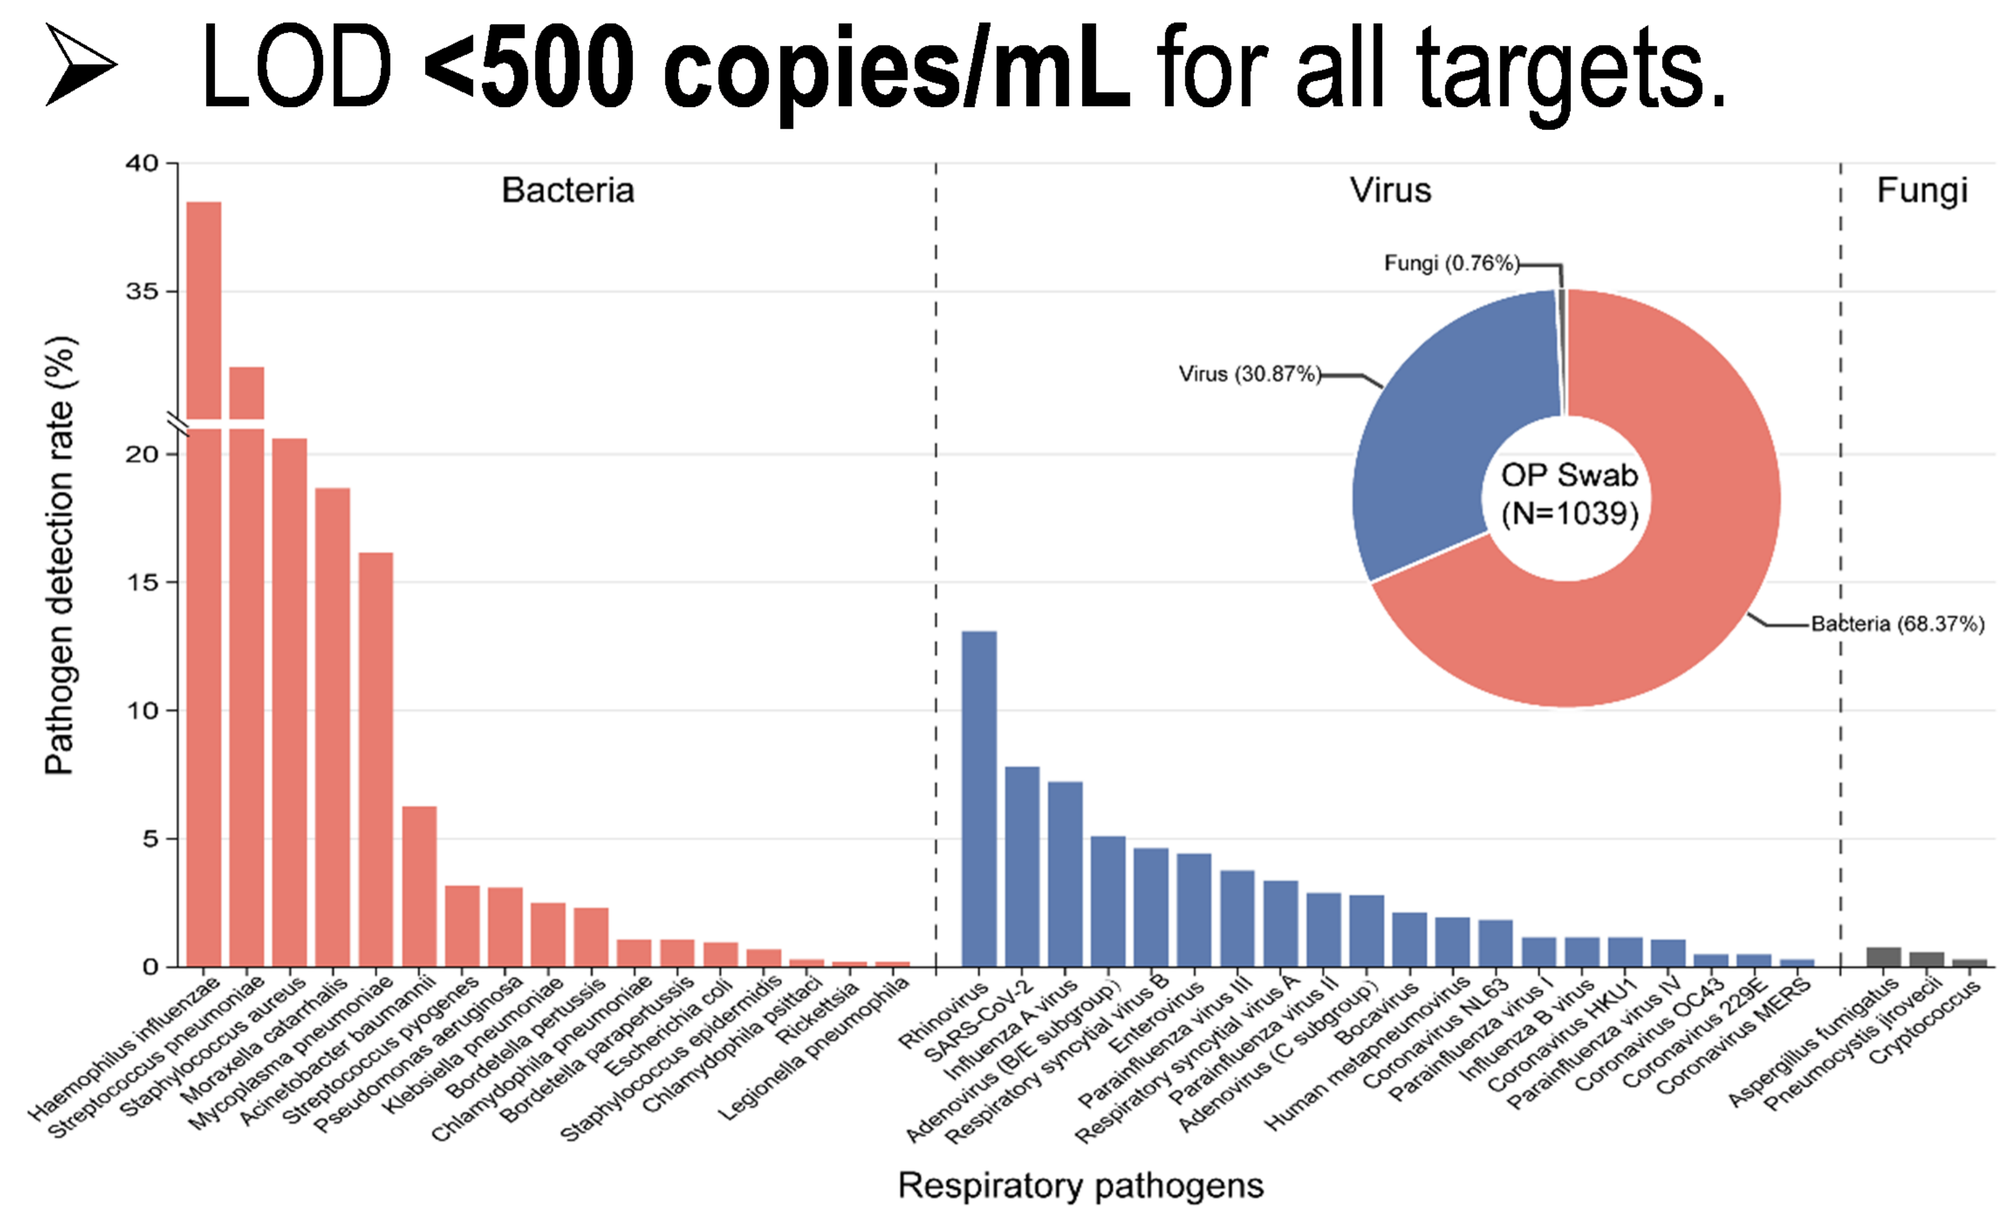
Bacteria
Virus
Fungi
Pathogen detection rate (%)
Fungi (0.76%)
Virus (30.87%)
OP Swab
(N=1039)
Bacteria (68.37%)
Respiratory pathogens

Figure 2. Clinical evaluation of RPP-40 in OP swab samples.

- Clinical performance analysis showed an overall concordance rate of 98.36–100% between RPP-40 and mNGS.

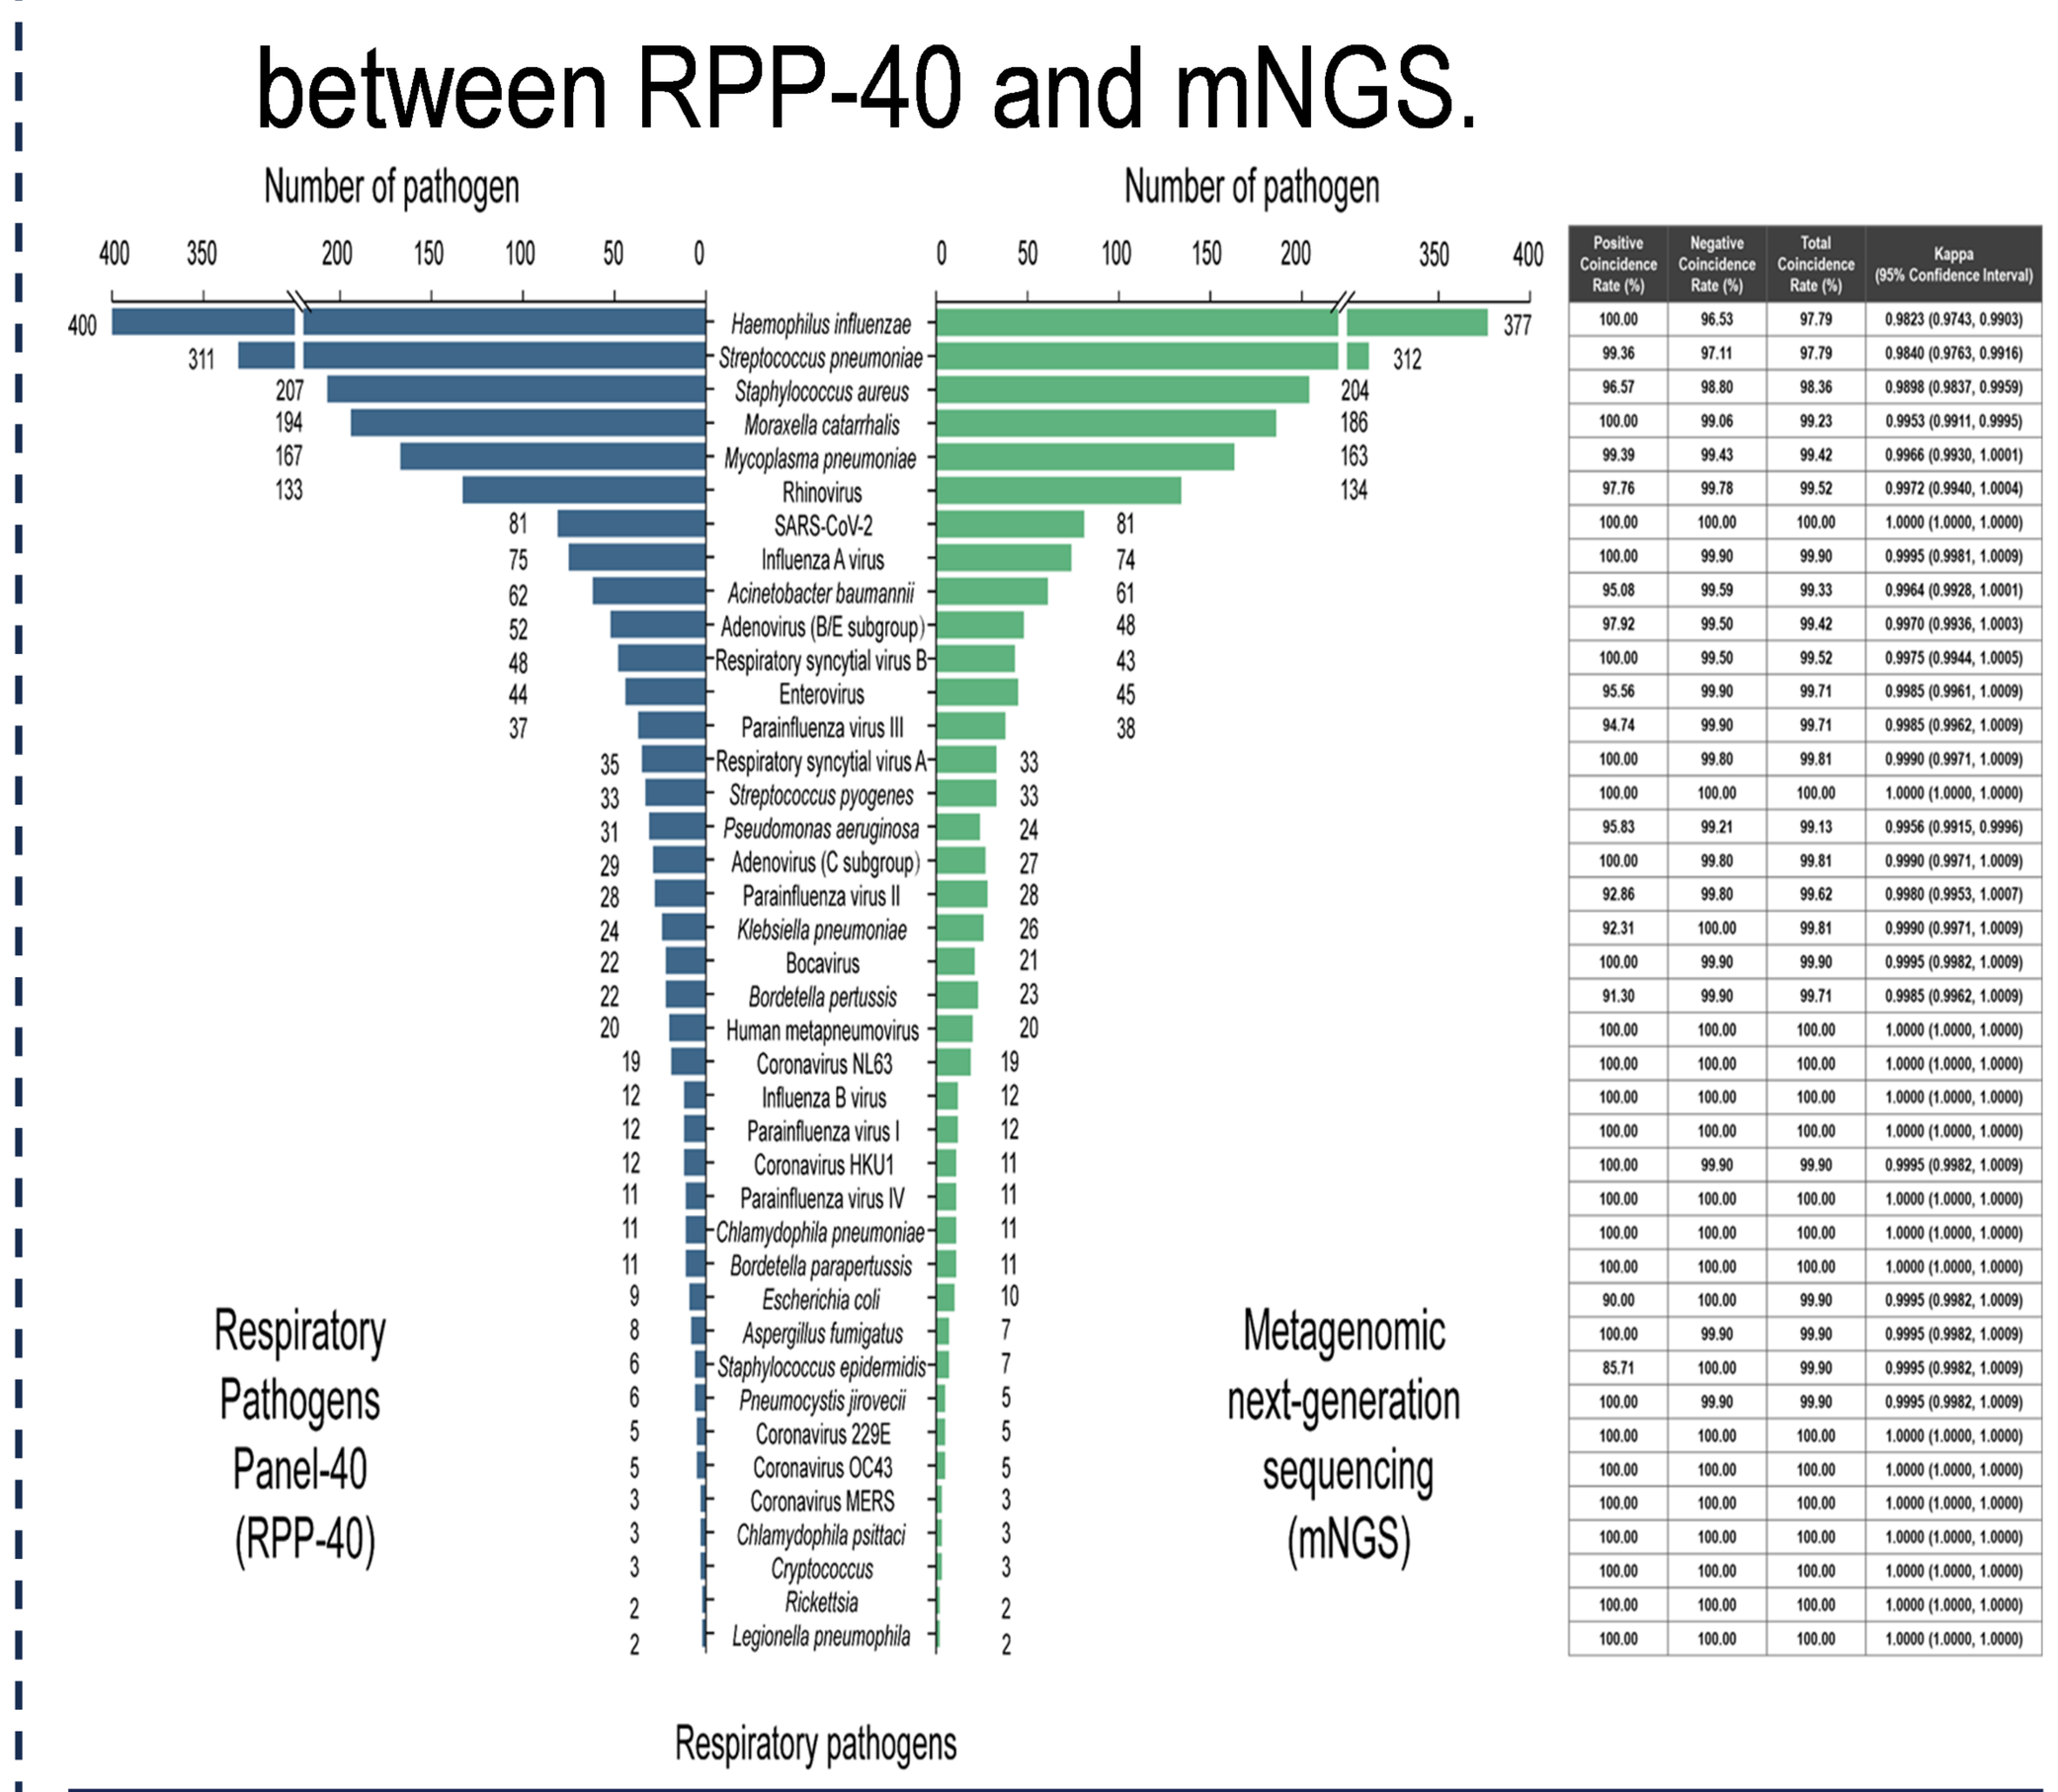
Number of pathogen
Number of pathogen
Respiratory
Pathogens
Panel-40
(RPP-40)
Metagenomic
next-generation
sequencing
(mNGS)
Positive Coincidence Rate (%)
Negative Coincidence Rate (%)
Total Coincidence Rate (%)
Kappa (95% Confidence Interval)
Respiratory pathogens

Figure 3. Comparative analysis of test results between RPP-40 and mNGS.

CONC.

The **RPP-40 assay** fills critical gaps in respiratory diagnostics by offering a robust, rapid, and scalable solution for comprehensive detection of respiratory pathogens. Its automation-ready design and high diagnostic accuracy make it well-suited for both routine clinical use and outbreak-response scenarios, aligning with global priorities for enhanced RTI management and evidence-based antimicrobial prescribing.

References: [1]. Kyu *et al. Lancet Infect Dis* 2025; [2]. Safiri *et al. Front Public Health* 2023; [3]. Huang *et al. PNAS* 2022.

Scan Me

APCCMI2025 BANGKOK

ACKNOWLEDGEMENTS: This study was supported by the NSFC (Grant No. 82472368).

CONTACT: Li Qingge, PhD, Tel. 86-0592-2187363; Fax. 86-0592-2187363; Email: qgli@xmu.edu.cn

ZEESAN Biotech 致善生物